The emotional impact of pyoderma gangrenosum (PG): Real-world experience of the PG patient journey

Afsaneh Alavi,[1] Dave De Pottie,[2] Valentina Dini,[3] Robert S. Kirsner,[4] Juan Semeco,[5] Alex G. Ortega-Loayza[6]

1. Department of Dermatology, Mayo Clinic, Rochester, Minnesota, USA; 2. Board Chair, All Things PG, Portland, Oregon, USA; 3. Dermatology Unit, Department of Clinical and Experimental Medicine, Ospedale Santa Chiara, Pisa, Italy; 4. Dr. Phillip Frost Department of Dermatology and Cutaneous Surgery, University of Miami Miller School of Medicine, Miami, Florida, USA; 5. Almirall S.A., Barcelona, Spain; 6. Department of Dermatology, Oregon Health and Science University, Portland, Oregon, USA.

Conclusions

This study emphasizes the substantial emotional impact of PG on study patients and caregivers, and highlights their challenges throughout the patient journey.

Addressing the core issue of delayed or misdiagnosis through improved disease education, and offering mental health and peer support, are crucial to empowering patients and improving their emotional and clinical outcomes.

Scan to download a copy of this poster
Copies of this poster and its content, obtained through this QR code, are for personal use only and may not be reproduced without written permission from the authors

Objective

- This qualitative research study explored the PG patient journey, with a focus on the emotional burden at multiple disease stages, and the critical need to provide more support to patients with PG.

Introduction

- PG is a rare, painful, ulcerative neutrophilic disorder that significantly impacts patients' physical, emotional, and psychosocial well-being.[1,2]
- However, due to the rarity of PG, data describing the emotional challenges faced by patients and their caregivers are limited.

Methods

- From August to September 2024, adults with PG and their caregivers were recruited through several routes, including:

PAGs and virtual panel

Referrals from HCPs

Diagnosis by an HCP was confirmed through a pre-screening questionnaire

Participants completed tasks on an online platform followed by either:

An online 60-minute individual interview

A 90-minute interview involving 2 patients

An independent market research agency conducted the interviews and collated the data

During the interviews, participants were asked about their experiences across **different stages of their PG journey**, with a particular focus on the **emotional and psychosocial impact**

Poster presented at the 2026 Winter Clinical Dermatology Conference – Hawaii, January 16–21, 2026, Maui, HI, USA.
Previously presented at the 2025 European Academy of Dermatology and Venereology Congress, September 17–20, 2025, Paris, France; the 2025 Fall Clinical Dermatology Conference, October 23–26, 2025, Las Vegas, NV, USA; the 2025 Innovations in Wound Healing Symposium, December 11–14, 2025, Key West, FL, USA; and the 2026 Maui Derm Caribbean Congress, January 7–10, 2026, San Juan, Puerto Rico.

Results

Baseline characteristics

Participants (N=26)

Patients with PG (n=22)

Caregivers (n=4)

Identified through:

- USA: *Mix of PAGs, foundations, and panel*
- China: *HCP referral*
- Japan: *Panel and blog*
- France: *HCP referral*

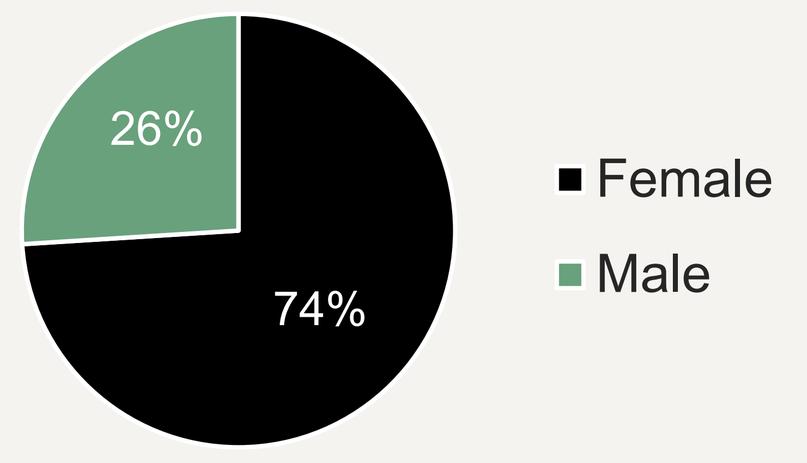
26%
74%
Female
Male

Overall, 74% of participants were female

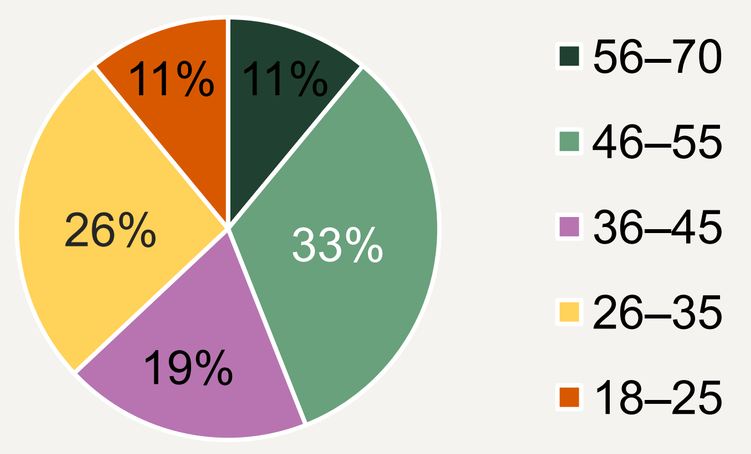
11%
11%
33%
19%
26%
56–70
46–55
36–45
26–35
18–25

Most patients (78%) were aged 26–55 years

Time since diagnosis was 1–5 years for 70% of patients

Overall, 42% of patients had comorbid conditions

Most patients (91%) had moderate-to-severe PG*

*Disease severity was self-reported.

Onset of symptoms

PG initially presents as a 'small bump' that grows aggressively into an ulcer

PG ulcers cause excruciating **pain** for patients

As symptoms developed, patients reported feeling:

Worried · Anxious · Depressed · Low self-esteem · Isolated

Patient author insights

"PG negatively affects relationships with your spouse, family, and friends. I feel so much guilt that I can no longer play with my grandkids or participate in family functions. I'm not the person I used to be."

Presentation

Patients with PG reported experiencing:

- High levels of distress due to size of ulcer
- Severe pain
- Immobilization
- Difficulty performing daily tasks

Patients with PG explained that PG **takes over their lives**, affecting their:

Routines · Ability to partake in hobbies · Wardrobe · Ability to socialize · Relationships with family and friends

Comorbid conditions may cause **delays** in presenting to a dermatologist

Patient author insights

"Misdiagnosis and mistreatment allow wounds to grow and go deep, resulting in higher risk of infection, loss of mobility, and adding years to recovery. Early diagnosis is the key."

Diagnosis

HCPs can struggle to **diagnose** and **advise** patients due to a **lack of objective markers for diagnosis** and **information** on the disease, leading to feelings of **hopelessness**, **fear**, and **uncertainty** among patients

Patients are often **misdiagnosed**, and diagnosis can be **delayed**, leading to:

- Feelings of **frustration**, **mistrust**, and **neglect**
- **Anxiety** and **stress** over a lack of clear explanation
- A lasting **impact on optimism** towards treatment options

Upon learning that PG is a chronic condition, many patients feel **depressed, embarrassed**, **scared**, and **isolated**

Treatment

Symptoms can cause patients to stop working, leading to **financial issues**, which can **limit treatment options**

After treatment failure or relapse, patients reportedly felt **isolated** and **pessimistic**, with emotions fluctuating due to uncertainty of treatment success and the impact on quality of life

Patients expressed high levels of **doubt** and **hopelessness** in response to uncertainty surrounding the success of their treatment outcomes

Ongoing management

Pain continues throughout treatment until wound closure, which can be a **lengthy process**, causing the patient **distress**; even upon wound closure, **symptoms can remain**, such as flare-ups, and dry and itchy skin

Patient author insights

"The constant, intense pain of PG takes over your life, wears you down, and destroys your quality of life. PG leaves deep physical and emotional scars that may never heal."

Patients reported **anxiety** and a **fear** of the risk of **relapse**

Those who experienced relapse felt like they were **failing** and **losing control**

Relapses are often **unexpected** and can be very **distressing** for patients, who often need **emotional support** during this time

Abbreviations

HCP, healthcare professional; PAG, patient advocacy group; PG, pyoderma gangrenosum.

References

1. Ruocco E, et al. J Eur Acad Dermatol Venereol 2009; 23:1008–1017;
2. Sanchez-Melendez SN, et al. Exp Dermatol 2024; 3:e14876.

Data sharing statement

To ensure independent interpretation of clinical study results and enable authors to fulfill their role and obligations under the ICMJE criteria, Boehringer Ingelheim grants all external authors access to relevant clinical study data. In adherence with the Boehringer Ingelheim Policy on Transparency and Publication of Clinical Study Data, scientific and medical researchers can request access to clinical study data, typically, one year after the approval has been granted by major Regulatory Authorities or after termination of the development program. Researchers should use the https://vivli.org/ link to request access to study data and visit https://www.mystudywindow.com/msw/datasharing for further information.

Acknowledgments

This study was conducted by Healthcare Research Worldwide (HRW) and supported and funded by Boehringer Ingelheim and LEO Pharma A/S. We thank all participants for their contributions to study implementation. The authors meet criteria for authorship as recommended by the International Committee of Medical Journal Editors (ICMJE) and did not receive payment related to the development of this poster. Boehringer Ingelheim and LEO Pharma A/S were given the opportunity to review the poster for medical and scientific accuracy, as well as intellectual property considerations. Shreyal Maikoo, PhD, of Nucleus Global, an Inizio company, provided medical writing support, which was contracted and funded by Boehringer Ingelheim and LEO Pharma A/S.

Disclosures

AA is receiving research support from La Roche-Posay, the Hidradenitis Suppurativa Foundation, and the National Institutes of Health, and has received research support from the Canadian Dermatology Foundation (now terminated); is receiving honoraria for consulting or participation in advisory boards from AbbVie, Almirall, Avalo Therapeutics, Boehringer Ingelheim, Cantargia, Incyte, LEO Pharma, Navigator Medicines, Novartis, Sanofi, and UCB; was Principal Investigator in clinical trials for Boehringer Ingelheim and Processa Pharmaceuticals (paid to their institution; now terminated); serves on a data monitoring board for InflaRx, Almirall, and Zura Bio; and is a board member of the Hidradenitis Suppurativa Foundation. **AGOL** is a member of the Honorary Editorial Board of the American Journal of Clinical Dermatology and Associate Editor of Dermatology (Karger); is a consultant for Castle Biosciences, Clarivate, Corvus Pharmaceuticals, Eli Lilly, Genentech, Guidepoint, InflaRx, Otsuka, Sitala Bio Ltd, and TFS Health Science; is an advisory board member for Biotech, Boehringer Ingelheim, Bristol Myers Squibb, Janssen, and Sanofi; and is receiving research grants from AbbVie, Boehringer Ingelheim, Eli Lilly, Incyte, InflaRx, Janssen, National Institutes of Health (supported by NIH NIAMS R01 AR083110), and Pfizer. **JS** is an employee of Almirall S.A. and was an employee of Boehringer Ingelheim at the time of this study. **DDP**, **VD**, and **RSK** have nothing to declare.